Relapses during addiction treatment found after a change in urine drug testing methodologies[1]

Adam Rzetelny,[2] PhD and Adrienne Marcus,[3] PhD

Introduction: Definitive urine drug testing (UDT) may provide a more complete picture of a patient's drug-taking behaviors than presumptive UDT. Definitive UDT involves mass spectrometry, either LC-MS/MS or GCMS, and may not require a screen. In contrast, presumptive UDT, sometimes referred to as screen-first, involves immunoassay, such as point-of-care cups, and chemical analyzers, and may be followed by confirmation of positive results. Substance-use counselors have previously reported that switching from presumptive to definitive UDT helped identify relapses and impacted the quality of counseling, treatment planning, and patient outcomes. However, an objective assessment of new relapses potentially identified when switching UDT methodologies had yet to be undertaken.

Objectives: The aim of the present study was to use chart-review in an addiction-treatment setting to investigate newly identified relapses when switching UDT methodologies from presumptive to definitive LCMS. The hypothesis was that more new relapses would be discovered after the switch. A secondary aim was to examine how additional information about patient drug-taking behaviors was utilized in treatment.

Methods: After IRB approval, chart reviews were conducted at two outpatient centers of a large substance-use treatment program that tested patients 2 to 3 times weekly and had independently chosen to switch UDT methods from presumptive to definitive. Charts were included if they covered a patient for at least one month prior to and following this switch and contained the actual UDT laboratory reports or clear evidence of UDT results in the clinical notes. Thus, all patients were tested with both methodologies in a quasi pre-post manner. A relapse was defined by an unexpected UDT result that was inconsistent with prescribed treatment, either positive for illicit drugs or medications not prescribed, or negative for prescribed medications. Charts were also reviewed for information regarding patient demographics and how UDT information was applied in treatment.

Notes. LC-MS/MS: Liquid Chromatography with Tandem Mass Spectrometry; GCMS: Gas Chromatography with Mass Spectrometry; ETG: ethyl-glucoronide; ETS: ethyl-sulfate; 6MAM: 6-monoacetylmorphine; UDT: Urine Drug Test

[1]This research was supported by Lexington Recovery, Katonah NY
[2]Collegium Pharmaceuticals, Stoughton MA
[3]Lexington Recovery, Katonah NY

For questions, please email Adam Rzetelny at adamiz@yahoo.com

Results: Forty-one charts met inclusion criteria, representing diverse demographics and substance-use histories.

Demographics: This sample was 56% male, and the average age was 35.9 years. Twenty-eight charts identified the patient as white, 3 as African American, and 5 as Hispanic. Ethnicity was unavailable in 6 charts. The primary diagnosis was opioid-use disorder (OUD; n=23), but all charts reflected polysubstance-use histories. Twenty-seven charts indicated that the patient was court-mandated to treatment; 3 were not mandated, and this information could not be found in 11 of the charts.

Relapses. Consistent with the hypothesis, of the 41 charts, 12 (29%) revealed relapses that were identified after the switch from presumptive to definitive UDT. Unexpected drug use newly identified by definitive testing included opioids, alcohol (ETS/ETG), and benzodiazepines. Definitive testing also revealed several cases of probable sample tampering (dilution). Some of these relapses were addressed and helped shape the course of treatment accordingly, and possible missed opportunities were also identified.

Relapse dispositions: In 9 of the 12 newly identified relapses, the chart indicated that the relapse was address by clinical staff by confronting the patient and intensifying treatment. In 4 of these cases, the patient was reported to attain abstinence and complete the program. In 5 of these cases, the relapse was addressed but the patient needed to be discharged to a higher level of care. In 2 of the 12 cases the relapse was not addressed, and in 1 of 12 relapses the disposition could not be determined by reviewing the chart.

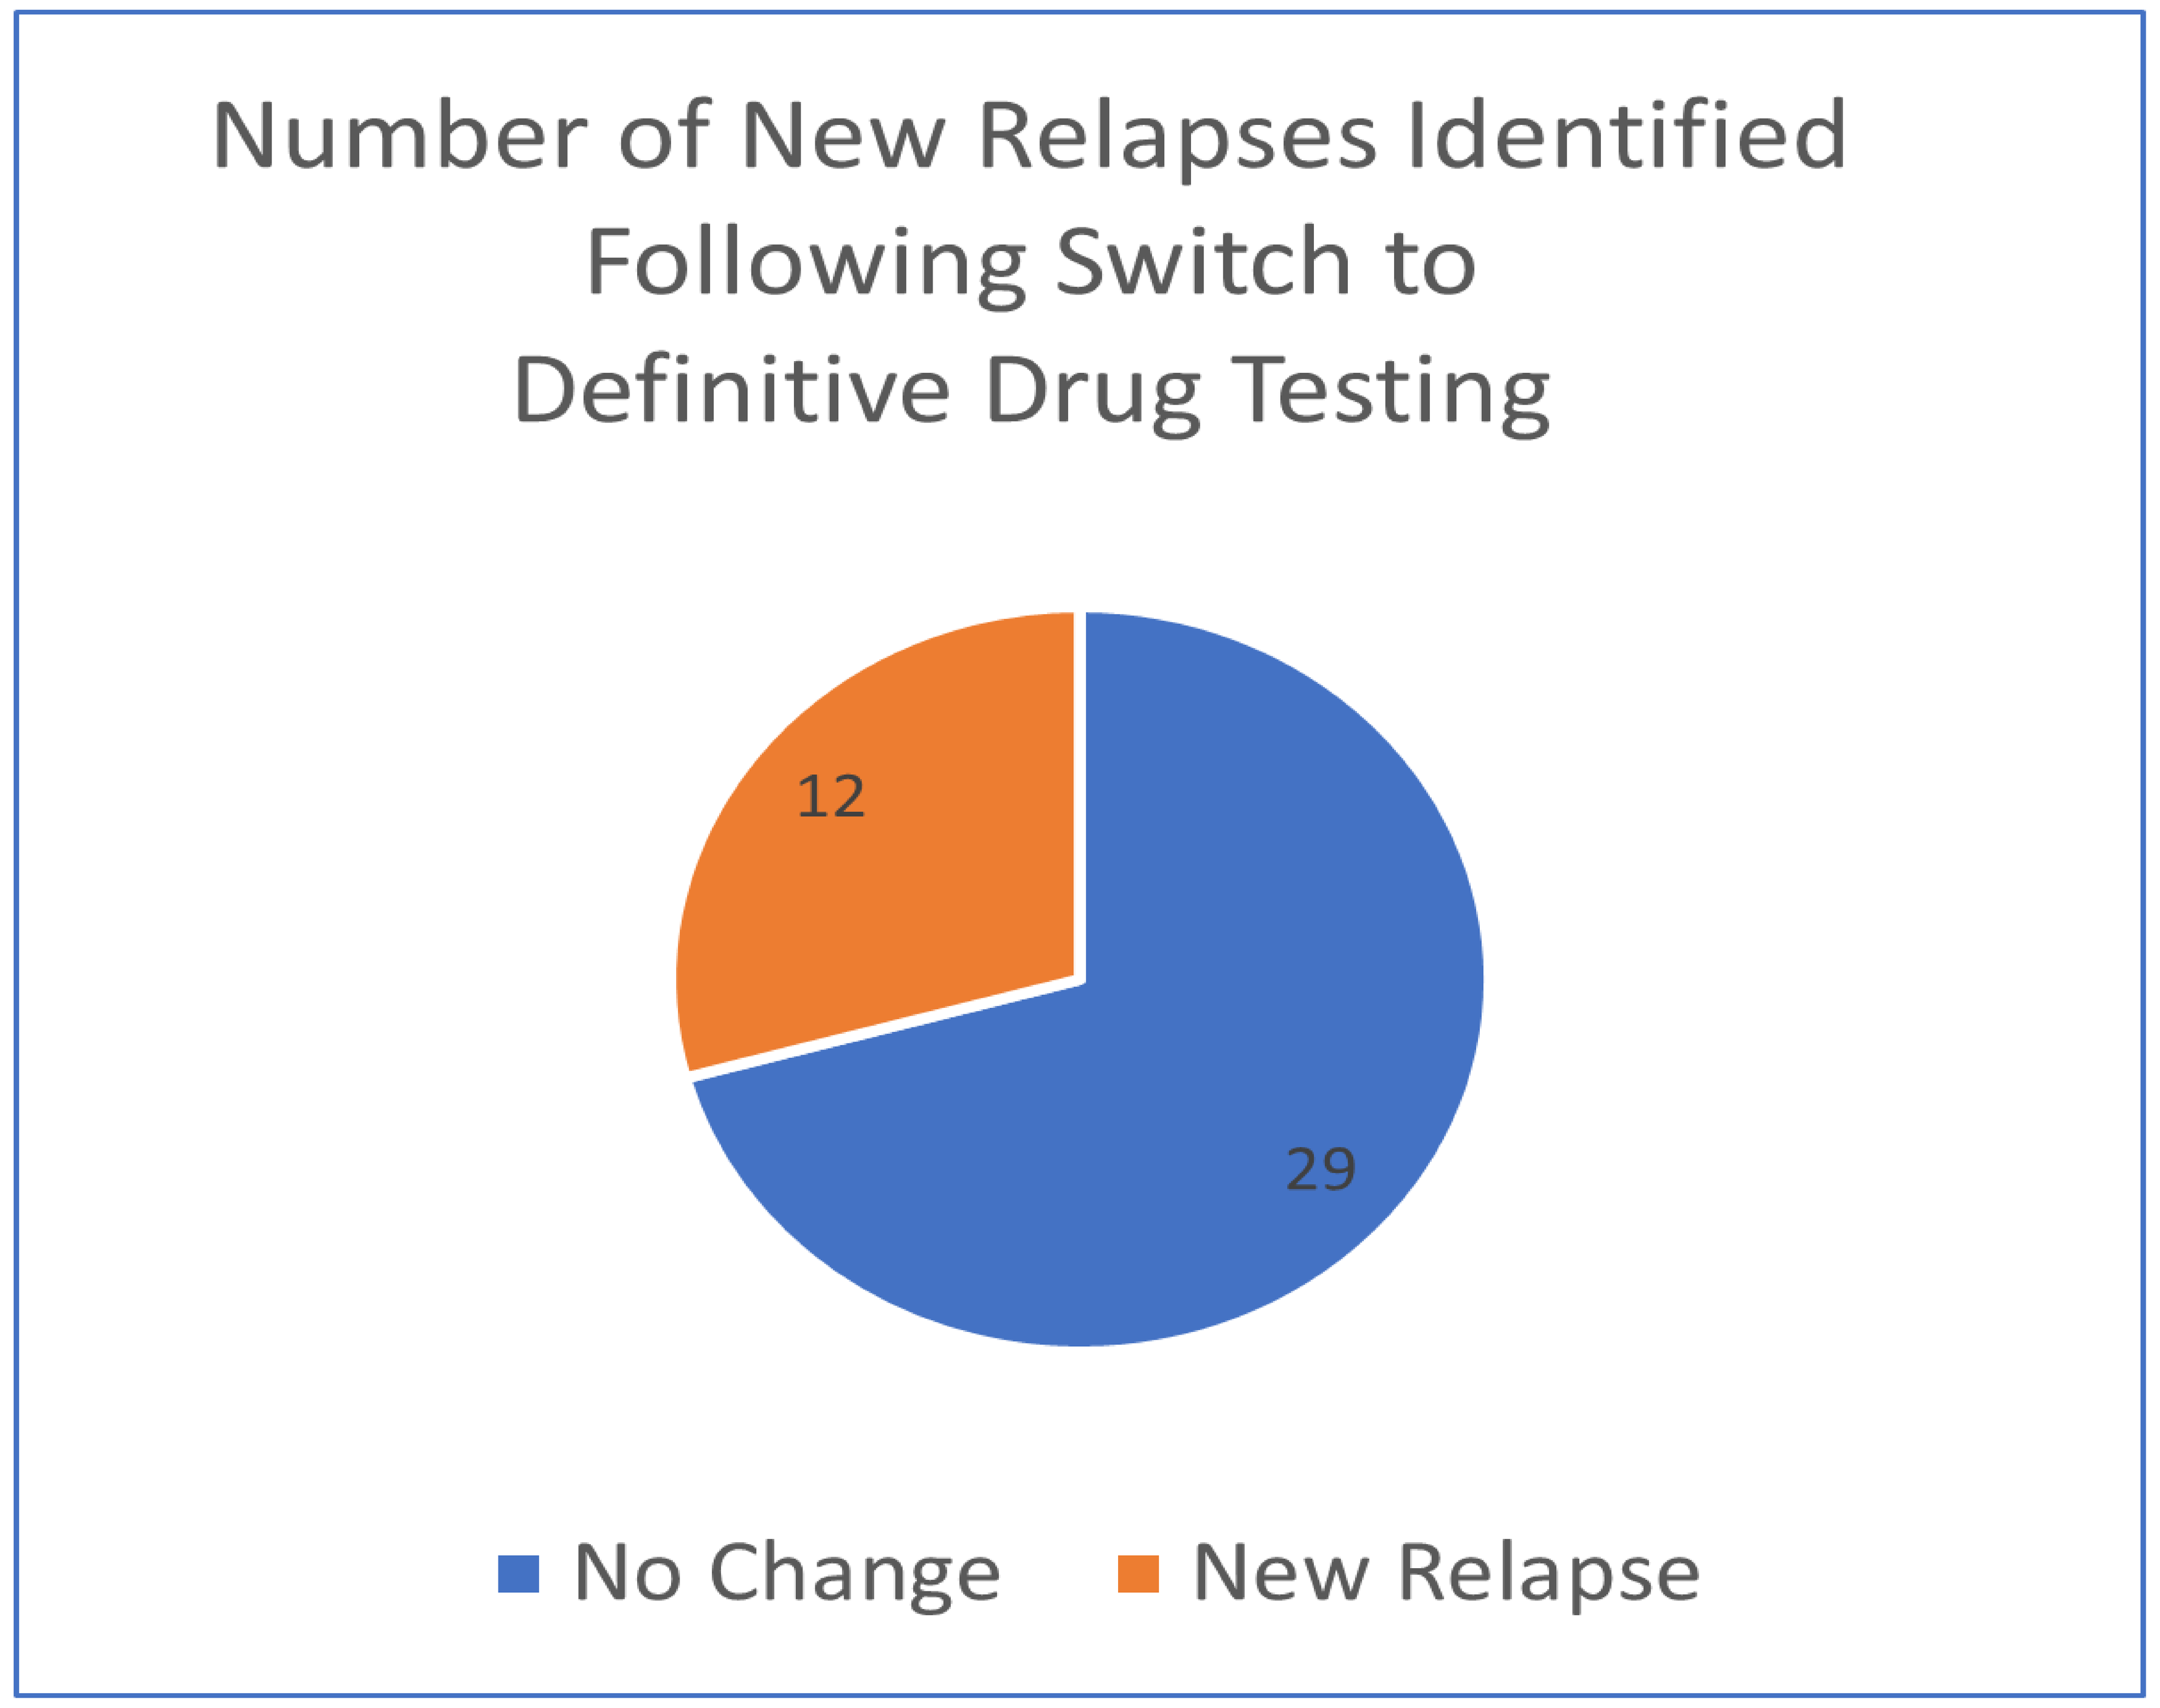
Number of New Relapses Identified Following Switch to Definitive Drug Testing
12
29
No Change
New Relapse

Selected relapse details. Of the 12 relapses newly identified after the introduction of definitive testing, 2 revealed that alprazolam (Xanax) use without prescription in place of the patient’s prescribed clonazepam (Klonopin). Prior to introducing definitive testing in this case, the patient’s presumptive results were positive for benzodiazepines but mistakenly interpreted as consistent with prescribed clonazepam adherence. In once case, the relapse was identified using definitive oral fluid rather than urine as the testing matrix because the patient was suspected by clinical staff of substituting urine samples in order to dissimulate, which was determined to be the case. In another case, results of presumptive tests were positive for opiates, which were assumed to be false positives by the staff until definitive testing revealed morphine and 6MAM, the latter being the definitive biomarker for heroin use. in yet another case, following the switch to definitive testing, buprenorphine “shaving” was revealed by missing metabolites and extraordinarily high quantities of the parent compound. Several cases of possible attempts to dilute the urine sample were identified by the investigator but not addressed by the clinical staff.

Conclusions: Switching from presumptive to definitive UDT revealed newly discovered relapses in 29% of these substance-use charts. This is an important finding considering that UDT is used to monitor a patient's progress in treatment in order to help guide treatment and optimize patient outcomes. Patients erroneously believed to be abstinent may receive a reduced level of care, potentially increasing their risk. This chart review also revealed opportunities to potentially improve how the results of UDT are utilized in practice and highlight areas for education on the clinical use of UDT in substance-use treatment, particularly around the potential attempts to dilute samples.

References:

- Rzetelny A, Zeller B, Miller N, Kirsh K, Passik S (2016). Definitive LC-MS/MS drug monitoring impacts substance-use treatment planning and patient outcomes: A brief report. J Addict Med Nov/Dec;10(6):443-447. PMID: 27649263.
- Rzetelny A, Zeller B, Miller N, Ruehle M, City KE, Kirsh KL, Passik SD (2016). Counselors’ clinical use of definitive drug testing results in their work with substance-use clients: A qualitative study. International Journal of Mental Health and Addiction, 14(1), 64-80. PMID: 26798328
- Kirsh KL, Baxter LE, Rzetelny A, et al. A survey of ASAM members’ knowledge, attitudes, and practices in urine drug testing. J Addict Med 2015;9:399–404.
- American Society of Addiction Medicine (ASAM). Drug Testing: A White Paper of the American Society of Addiction Medicine 2013.